SRK-181, A Latent TGFβ1 inhibitor: Safety, Efficacy, and Biomarker Results from the Dose Escalation Portion of a Phase 1 Trial (DRAGON Trial) in Patients with Advanced Solid Tumors

Timothy A. Yap[1], Justin F. Gainor[2], Meredith McKean[3], Melissa Johnson[3], Bruno Bockorny[4], Minal Barve[5], Randy F. Sweis[6], Ulka Vaishampayan[7], Ahmad Tarhini[8], Deepak Kilari[9], Yawen Ju[10], Si-Tuen Lee-Hoeflich[10], Stephen DeWall[10], Lan Liu[10], Nisha Shah[10], Ann Marie Kennedy[10], Lu Gan[10]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Massachusetts General Hospital Harvard Medical School, Boston, MA; [3]Sarah Cannon Research Institute, Nashville, TN; [4]Beth Israel Deaconess Medical Center, Boston, MA; [5]Mary Crowley Cancer Research, Dallas, TX; [6]University of Chicago, Chicago, IL; [7]University of Michigan, Ann Arbor, MI; [8]Moffitt Cancer Center, Tampa, FL; [9]Medical College of Wisconsin, Milwaukee, WI; [10]Scholar Rock, Inc., Cambridge, MA

Background

- Although many patients benefit from CPI therapies, the high incidence of drug resistance to CPI remains a challenge.[1]
- TGFβ1 plays an important role in mediating the primary resistance to PD-(L)1 blockade.[2-4]
- TGFβ1 is expressed in a latent form that needs to be activated for receptor binding.[4]
- SRK-181 is an investigational, fully human, selective anti-latent TGFβ1 IgG4 monoclonal antibody.[4]
- Preclinical results from a mouse tumor model (MBT-2) suggest that circulatory TGFβ1 may be a potential pharmacodynamic biomarker of SRK-181.[5]
- Compared to broad TGFβ inhibitors, SRK-181 demonstrates an improved safety profile in GLP nonclinical toxicology studies with no cardiotoxicities.[4,6-9]
- SRK-181 may improve upon nonselective TGFβ pathway approaches by decreasing PD-(L)1 inhibitor resistance and toxicity in cancer patients.

TGFβ1 Blockade with SRK-181 Renders Mouse Tumor Models Susceptible to Anti-PD-1 Therapy and Increases CD8+ T Cells Significantly in Tumors[4]

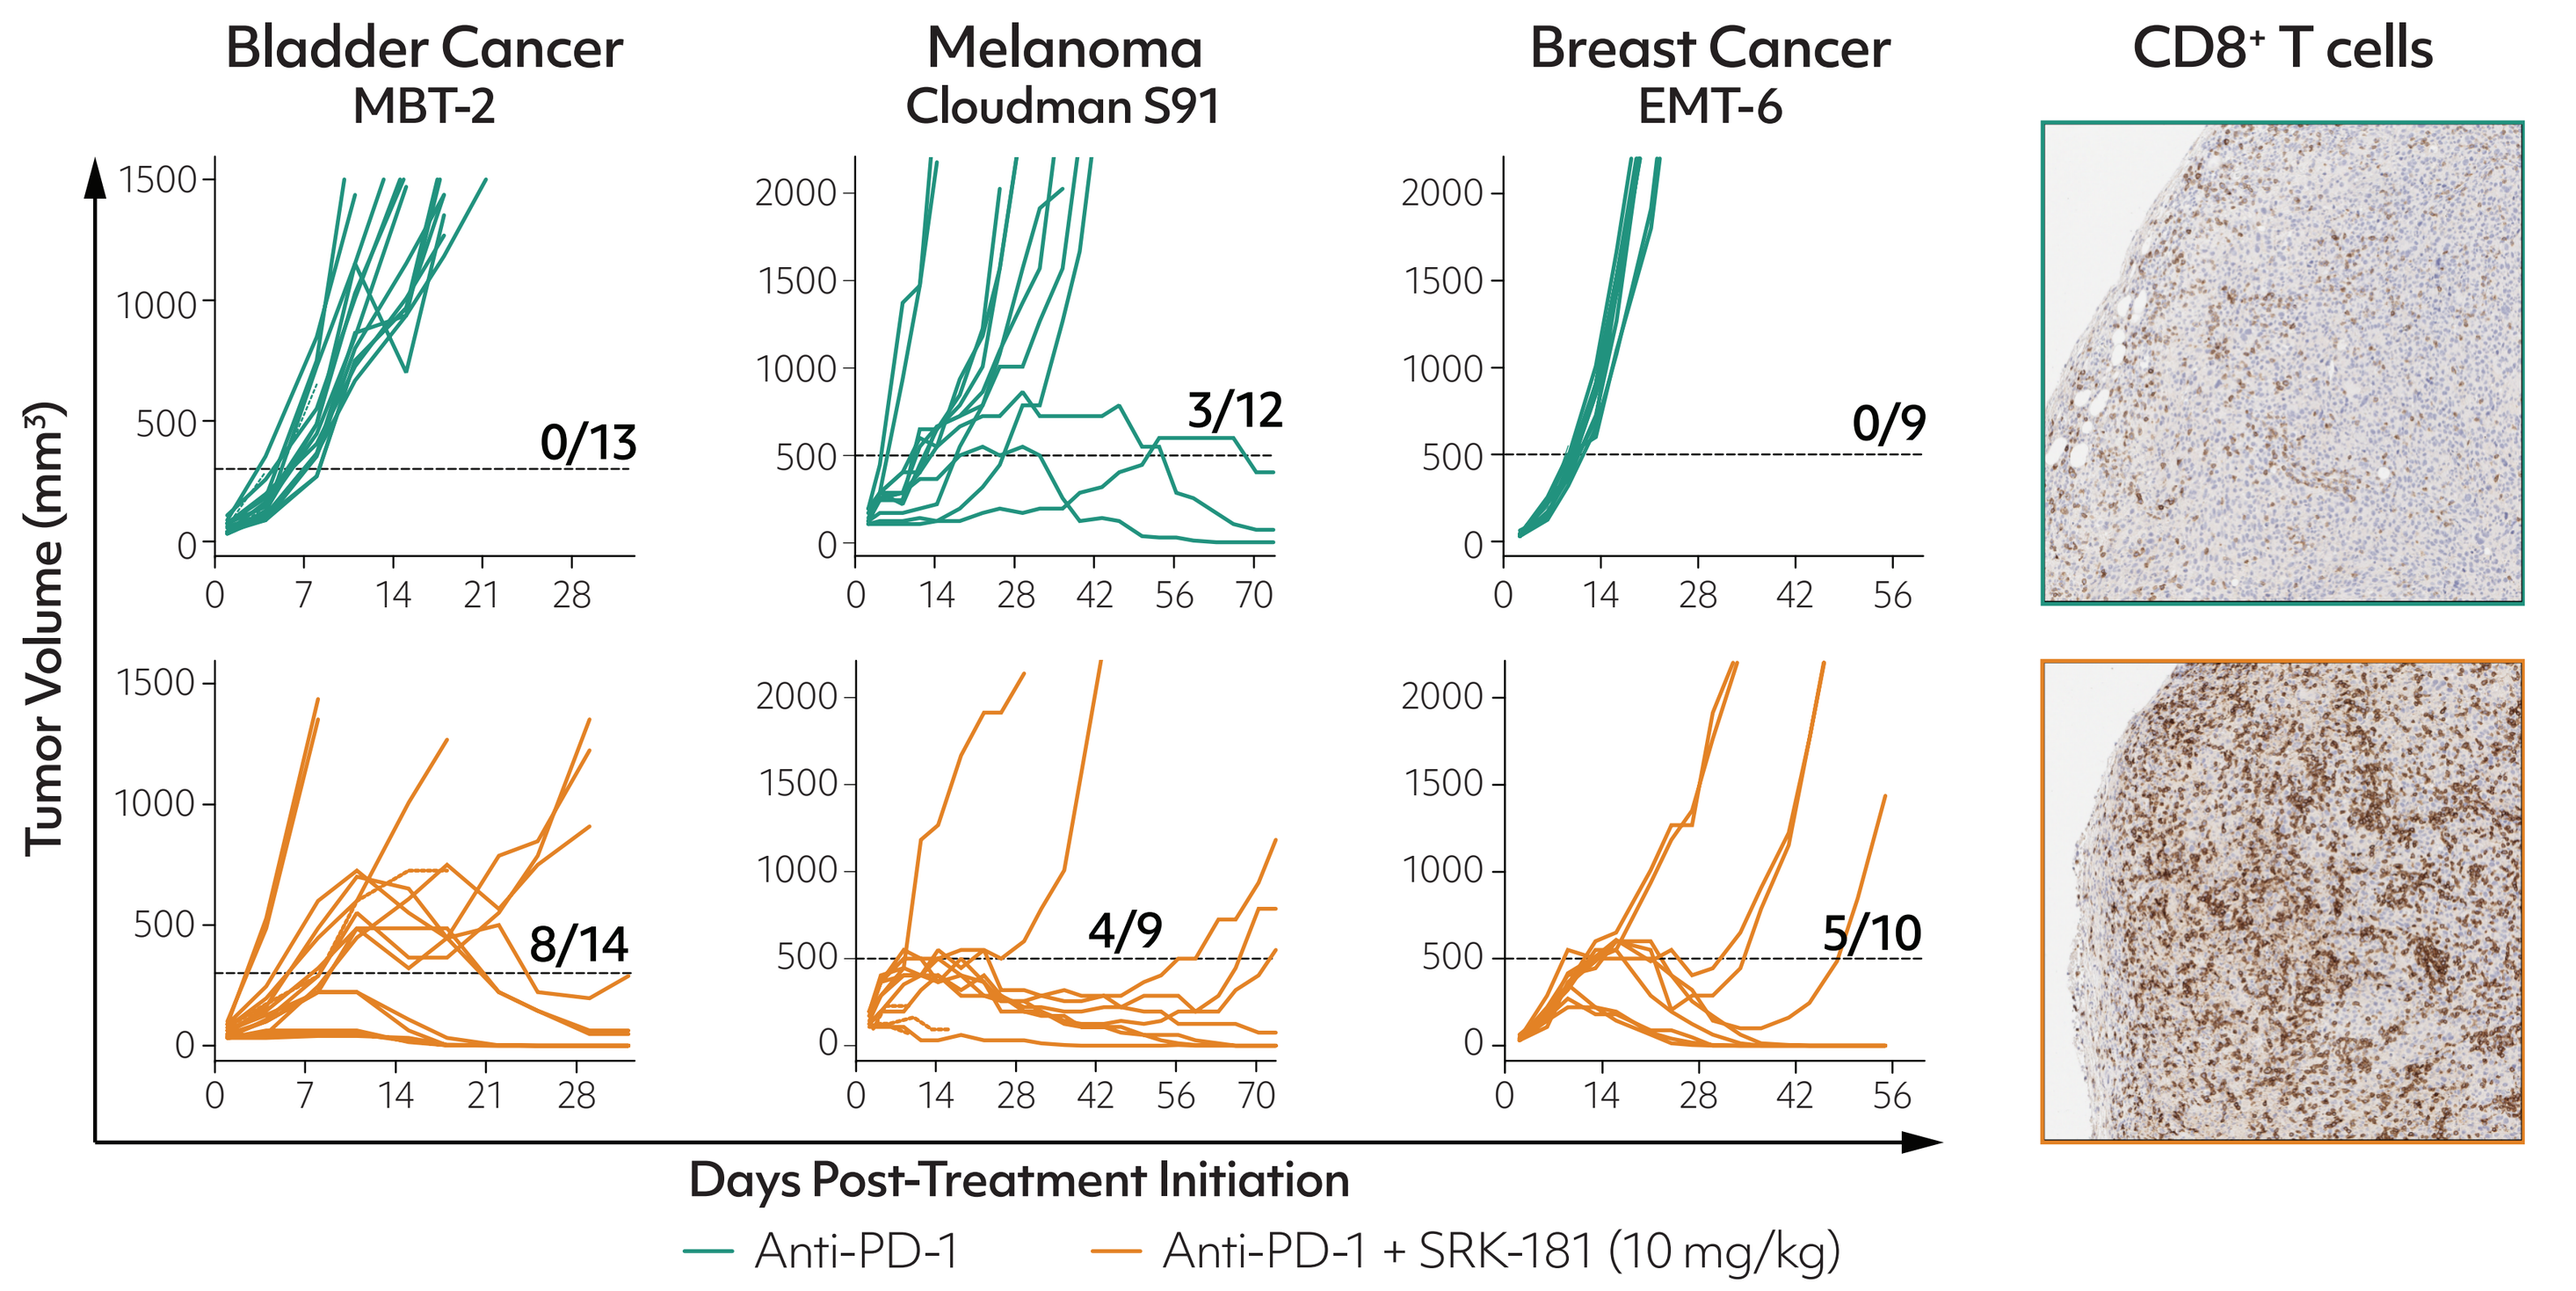
Bladder Cancer
MBT-2
Melanoma
Cloudman S91
Breast Cancer
EMT-6
CD8+ T cells
Tumor Volume (mm³)
0/13
3/12
0/9
8/14
4/9
5/10
Days Post-Treatment Initiation
Anti-PD-1
Anti-PD-1 + SRK-181 (10 mg/kg)

Phase 1 Clinical Study Overview

The DRAGON trial (NCT04291079) is a multicenter, open-label, Phase 1, FIH, dose-escalation, and dose-expansion trial to evaluate the safety, tolerability, PK, pharmacodynamics, and efficacy of SRK-181 alone and in combination with anti-PD-(L)1 therapy in patients with advanced solid tumors.

Part A: Dose Escalation (3 + 3 Design)

Part A1: SRK-181 (IV)

Advanced solid tumors that failed SOC treatment (completed)

- SRK-181 80 mg q3w (n = 1)
- SRK-181 240 mg q3w (n = 1)
- SRK-181 800 mg q3w (n = 3)
- SRK-181 1600 mg q3w (n = 4)
- SRK-181 2400 mg q3w (n = 3)
- SRK-181 3000 mg q3w (n = 3)
- SRK-181 2000 mg q2w (n = 4)*

Part A2 began

Part A2: SRK-181 (IV) + Anti-PD-(L)1

Non-Responders to prior Anti-PD-(L)1; Best response of SD/PD after 3 cycles (enrollment completed; 1 pt on treatment)

- SRK-181 240 mg q3w (n = 3)
- SRK-181 800 mg q3w (n = 3)
- SRK-181 1600 mg q3w (n = 6)
- SRK-181 2400 mg q3w (n = 3)

Part A Endpoints

Primary:
- Safety and tolerability (incidence/severity of AEs and SAEs)
- MTD or MAD
- Recommended dose for Part B

Secondary:
- PK
- ADA

Exploratory:
- Anti-tumor activity (ORR, DOR)
- Biomarker

*2000 mg q2w allows for equivalent C_{ave} exposure to 3000 mg q3w and is being evaluated to understand safety and PK of the q2w dose regimen

Part B: Dose Expansion (n = up to 40 per cohort)

Part B: SRK-181 (IV) + Anti-PD-(L)1

(enrolling)

- Cohort NSCLC (non-responders to prior anti-PD-1)
- Cohort UC (non-responders to prior anti-PD-1)
- Cohort MEL (non-responders to prior anti-PD-1)
- Cohort ccRCC (disease progressed on the most recent prior anti-PD-1)
- Cohort Any Other (non-responders to prior anti-PD-[L]1)

Part B Endpoints

Primary:
- Safety and tolerability (incidence/severity of AEs and SAEs)

Secondary:
- Anti-tumor activity (ORR, DOR)
- PK
- ADA

Exploratory:
- Survival outcomes (PFS, OS)
- Biomarker

Demographics and Baseline Characteristics

- As of August 29, 2022, 34 patients have been dosed:
 - In Part A1, 19 patients were dosed
 - In Part A2, 15 patients were dosed

Category	Part A1	Part A2
N	19	15
Age, median (range)	66 (41-79)	65 (32-75)
Gender (F/M)	8 / 11	3 / 12
Ethnicity		
Hispanic or Latino	0	2
Not Hispanic or Latino	18	13
Not Reported	1	0
Race		
White	19	15
Prior Lines of Therapy, median (range)	4 (1, 10)	3 (2, 7)

Patient Disposition

Category	Part A1	Part A2
Enrolled	19	15
On Study	0	1
Stopped Treatment	19	14
Reason for Completion/Discontinuation		
Adverse Event	2*	4**
Clinical Progression	3	4
Investigator Decision	2	1
Disease Progression based on RECIST v1.1	11	3
Withdrawal of Consent	1	2

Clinical cutoff date: August 29, 2022

*In Part A1, 1 patient discontinued from study due to an AE of road traffic accident that was unrelated to SRK-181 treatment; 1 patient discontinued due to an SRK-181-related AE of rash maculo-papular.

**In Part A2, 2 patients discontinued from the study due to treatment-unrelated AEs of spinal cord compression and ascites; 1 patient discontinued due to an anti-PD-(L)1-related AE of rash maculo-papular; 1 patient discontinued due to an SRK-181-related AE of pemphigoid.

Safety

Treatment-Emergent AEs Related to SRK-181, All Grades > 10% (Part A1)

Dose (mg)	80 (n = 1)	240 (n = 1)	800 (n = 3)	1600 (n = 4)	2400 (n = 3)	3000 (n = 3)	2000 (n = 4)	All (n = 19)
Fatigue	0	1	0	0	1	0	1	3 (15.8%)
Decreased appetite	1	0	1	0	0	0	0	2 (10.5%)
Nausea	1	0	0	0	0	0	1	2 (10.5%)

All dose levels were administered q3w except 2000 mg, which was administered q2w

Clinical cutoff date: August 29, 2022

Treatment-Emergent AEs Related to SRK-181 or Anti-PD(L)1, All Grades > 10% (Part A2)

Dose (mg)	240 (n = 3)	800 (n = 3)	1600 (n = 6)	2400 (n = 3)	All (n = 15)
Pruritus	1	0	1	1	3 (20.0%)
Rash	0	1	0	2	3 (20.0%)
Rash maculo-papular	1	0	1	1	3 (20.0%)
Diarrhoea	0	0	2	0	2 (13.3%)

*All dose levels were administered q3w

Clinical cutoff date: August 29, 2022

- Treatment-related Grade 3 AEs were alanine aminotransferase increased (1 patient in Part A1); pruritus (2 patients in Part A2), blister, immune-mediated lung disease, rash and rash maculo-papular (1 patient each in Part A2).
- No Grade 4 or 5 treatment-related AEs occurred.
- A treatment-related SAE of elevated troponin I (1 patient) was observed in Part A1; blister, pruritus, and rash (all in 1 patient) and immune-mediated lung disease (1 patient) were observed in Part A2.
- No DLTs were observed up to 3000 mg q3w and 2000 mg q2w in Part A1 and up to 2400 mg q3w in Part A2.

Abbreviations: ADA, anti-drug antibody; AE, adverse event; Anti-PD-(L)1, programmed death ligand-1 antibody/programmed cell death protein-1 antibody; C_{avg}, average plasma concentration; ccRCC, clear cell renal cell carcinoma; CPI, checkpoint inhibitor; DOR, duration of response; DLT, dose-limiting toxicity; ELISA, enzyme-linked immunosorbent assay; FIH, first in human; GLP, good laboratory practice; MAD, maximum administered dose; MEL, melanoma; MTD, maximum tolerated dose; NSCLC, non-small cell lung cancer; ORR, overall response rate; OS, overall survival; PD, progressive disease; PF4, platelet factor 4; PFS, progression-free survival; PI, principal investigator; PK, pharmacokinetics; PR, partial response; q2w, every 2 weeks; q3w, every 3 weeks; RCC, renal cell carcinoma; SAE, serious adverse event; SD, stable disease; SOC, standard of care; $T_{1/2}$, haf-life; TGFβ1, transforming growth factor beta-1; TNBC, triple negative breast cancer; UC, urothelial carcinoma.

Preliminary Efficacy

- Among 19 patients in Part A1:
 - Eight patients had a best response of SD (3 ovarian, 3 colorectal, 1 pancreatic, 1 testicular cancer).
 - All three patients with ovarian cancer were stable beyond the 16 week cutoff (range: 25 - 42 weeks).
- Among 15 patients in Part A2:
 - One confirmed RECIST1.1 PR was observed at 800mg in a patient with anti-PD-1 resistant ccRCC who remained on study for 30 weeks.
 - Nine patients had a best response of SD (3 head and neck cancer, 2 melanoma, 1 liver cancer, 1 RCC, 1 squamous cell skin carcinoma, 1 TNBC).
 - Six patients (highlighted below) were stable beyond the 16 week cutoff (2 head and neck cancer, 2 RCC, 1 melanoma, 1 skin squamous cell carcinoma); one ongoing patient with head and neck cancer had a 29.4% tumor reduction.
 - All 6 patients were progressed on prior anti-PD-(L)1 treatment.
 - The 6 patients had 3-6 lines of prior treatment.

Duration of Treatment

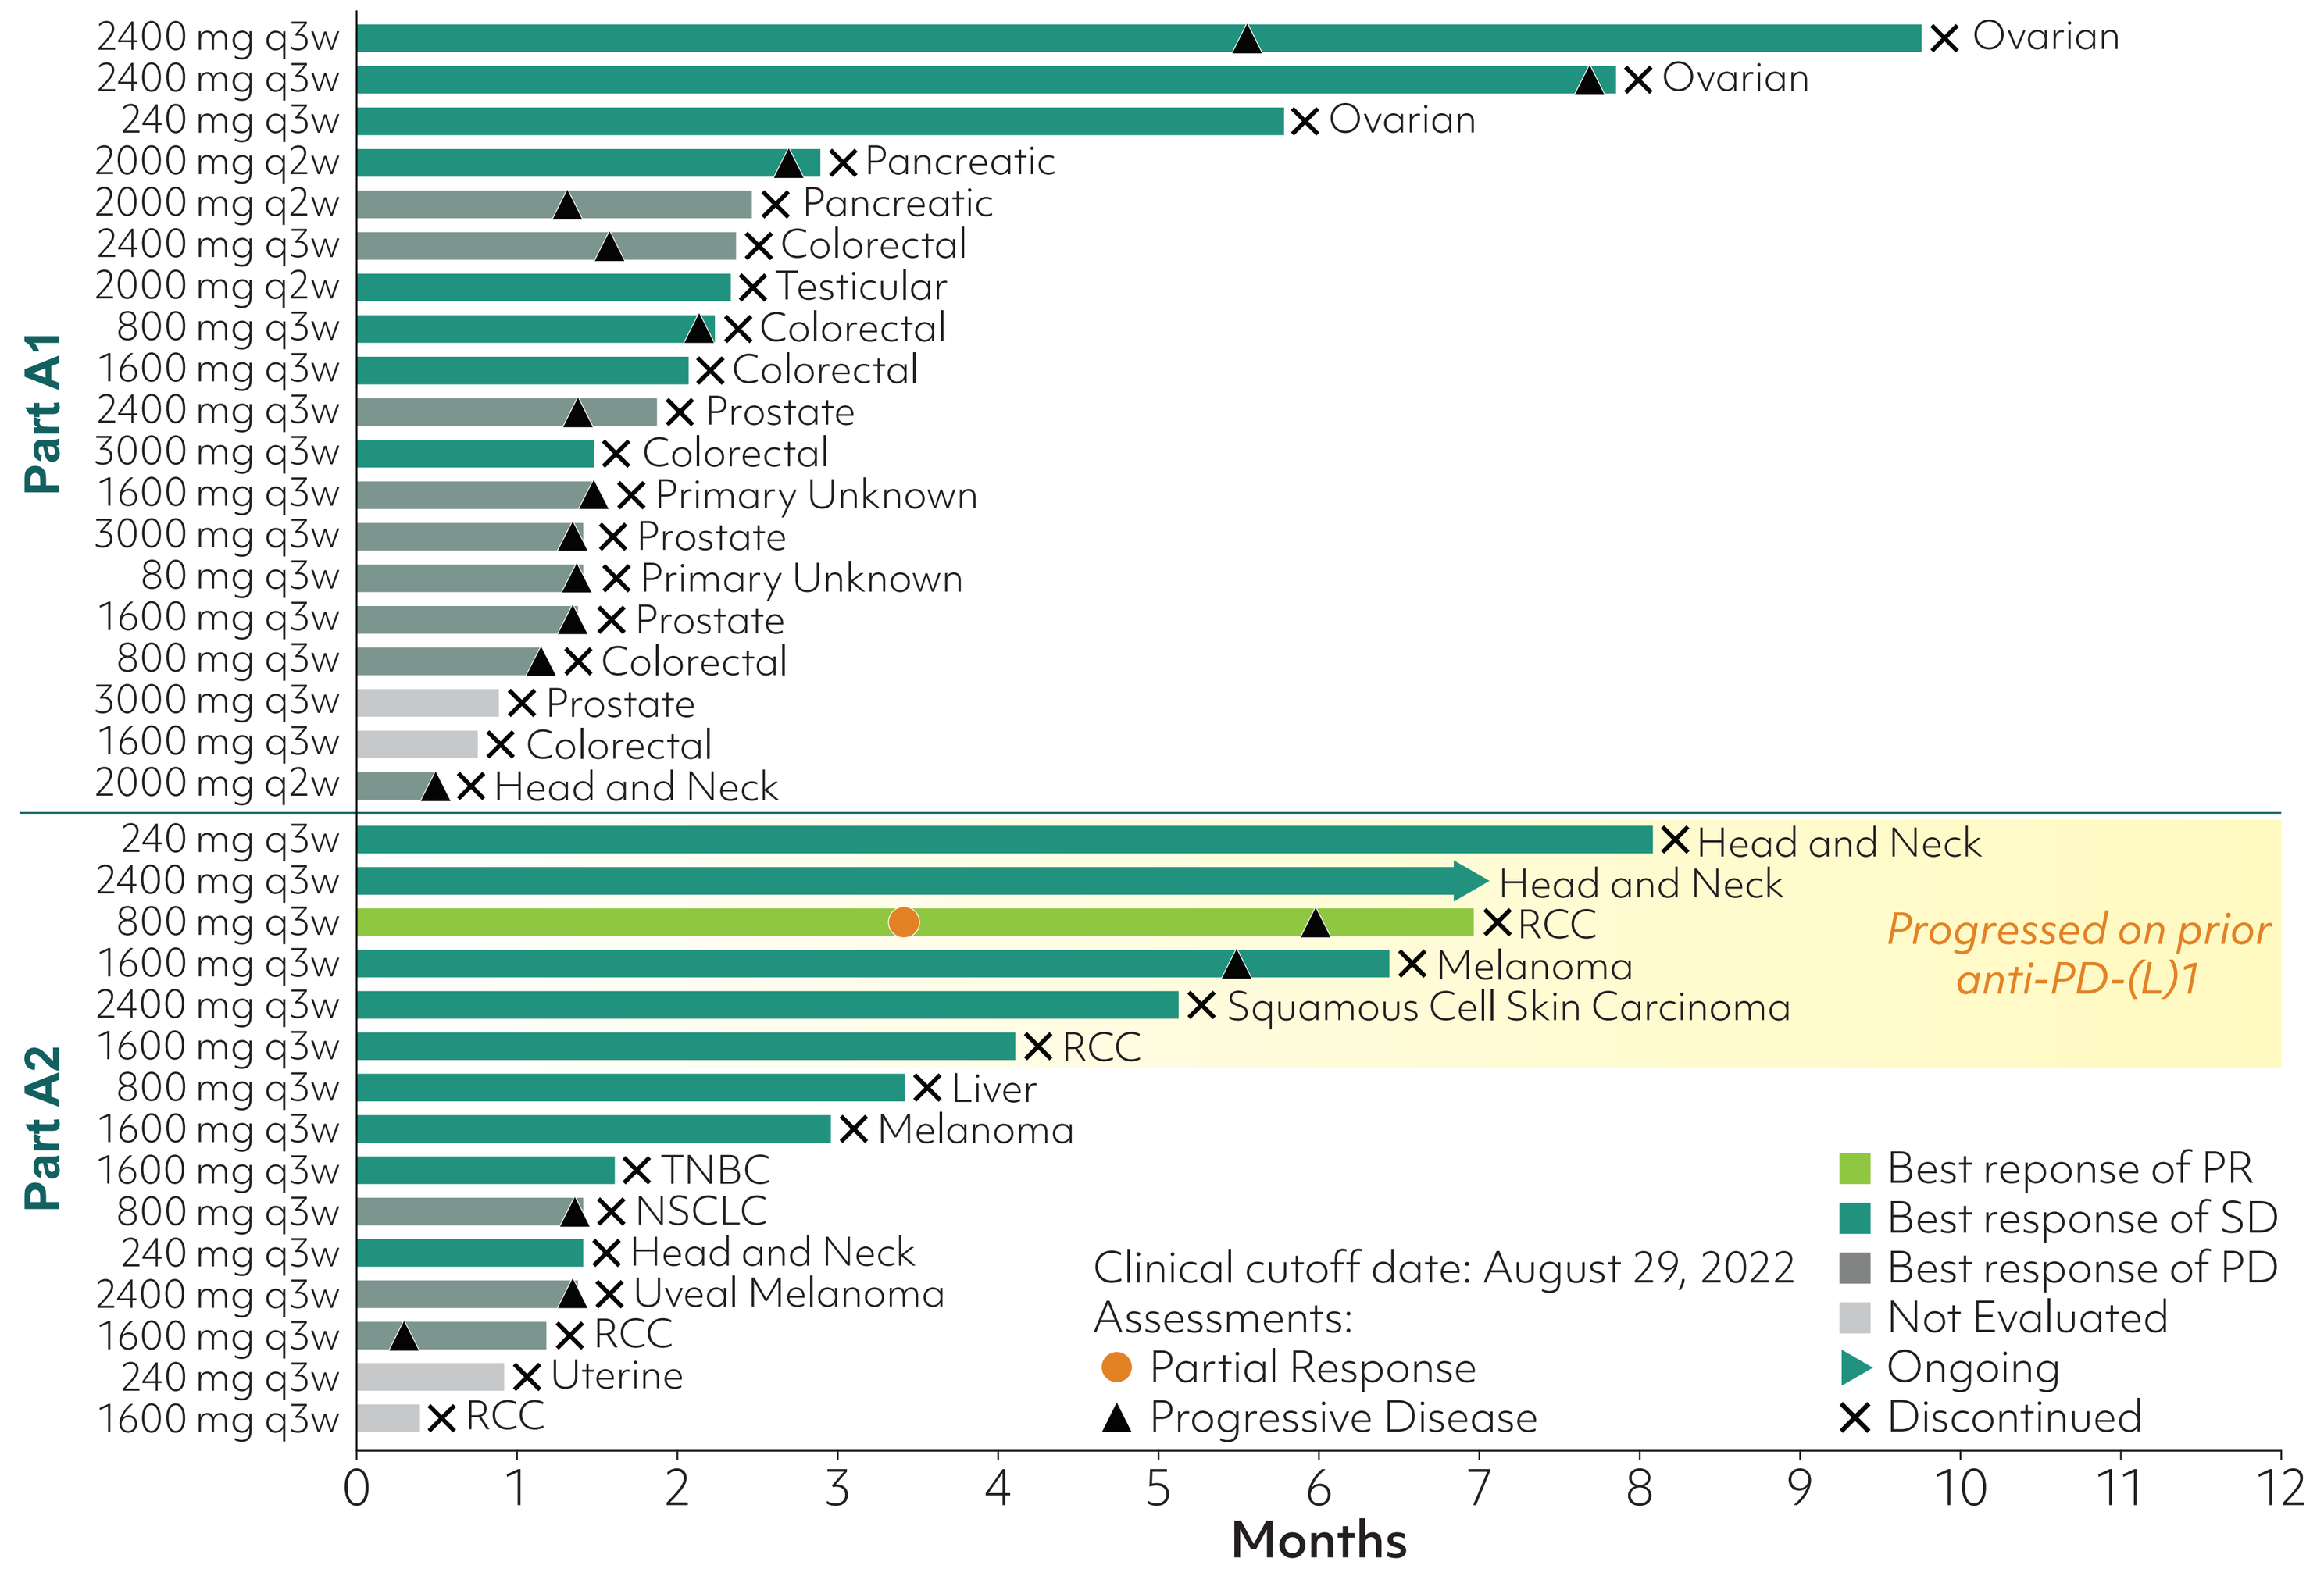
Part A1
2400 mg q3w Ovarian
2400 mg q3w Ovarian
240 mg q3w Ovarian
2000 mg q2w Pancreatic
2000 mg q2w Pancreatic
2400 mg q3w Colorectal
2000 mg q2w Testicular
800 mg q3w Colorectal
1600 mg q3w Colorectal
2400 mg q3w Prostate
3000 mg q3w Colorectal
1600 mg q3w Primary Unknown
3000 mg q3w Prostate
80 mg q3w Primary Unknown
1600 mg q3w Prostate
800 mg q3w Colorectal
3000 mg q3w Prostate
1600 mg q3w Colorectal
2000 mg q2w Head and Neck
Part A2
240 mg q3w Head and Neck
2400 mg q3w Head and Neck
800 mg q3w RCC
1600 mg q3w Melanoma
2400 mg q3w Squamous Cell Skin Carcinoma
1600 mg q3w RCC
800 mg q3w Liver
1600 mg q3w Melanoma
1600 mg q3w TNBC
800 mg q3w NSCLC
240 mg q3w Head and Neck
2400 mg q3w Uveal Melanoma
1600 mg q3w RCC
240 mg q3w Uterine
1600 mg q3w RCC
Progressed on prior anti-PD-(L)1
Best reponse of PR
Best response of SD
Best response of PD
Not Evaluated
Ongoing
Discontinued
Clinical cutoff date: August 29, 2022
Assessments:
Partial Response
Progressive Disease
Months

Best Response in Target Lesions

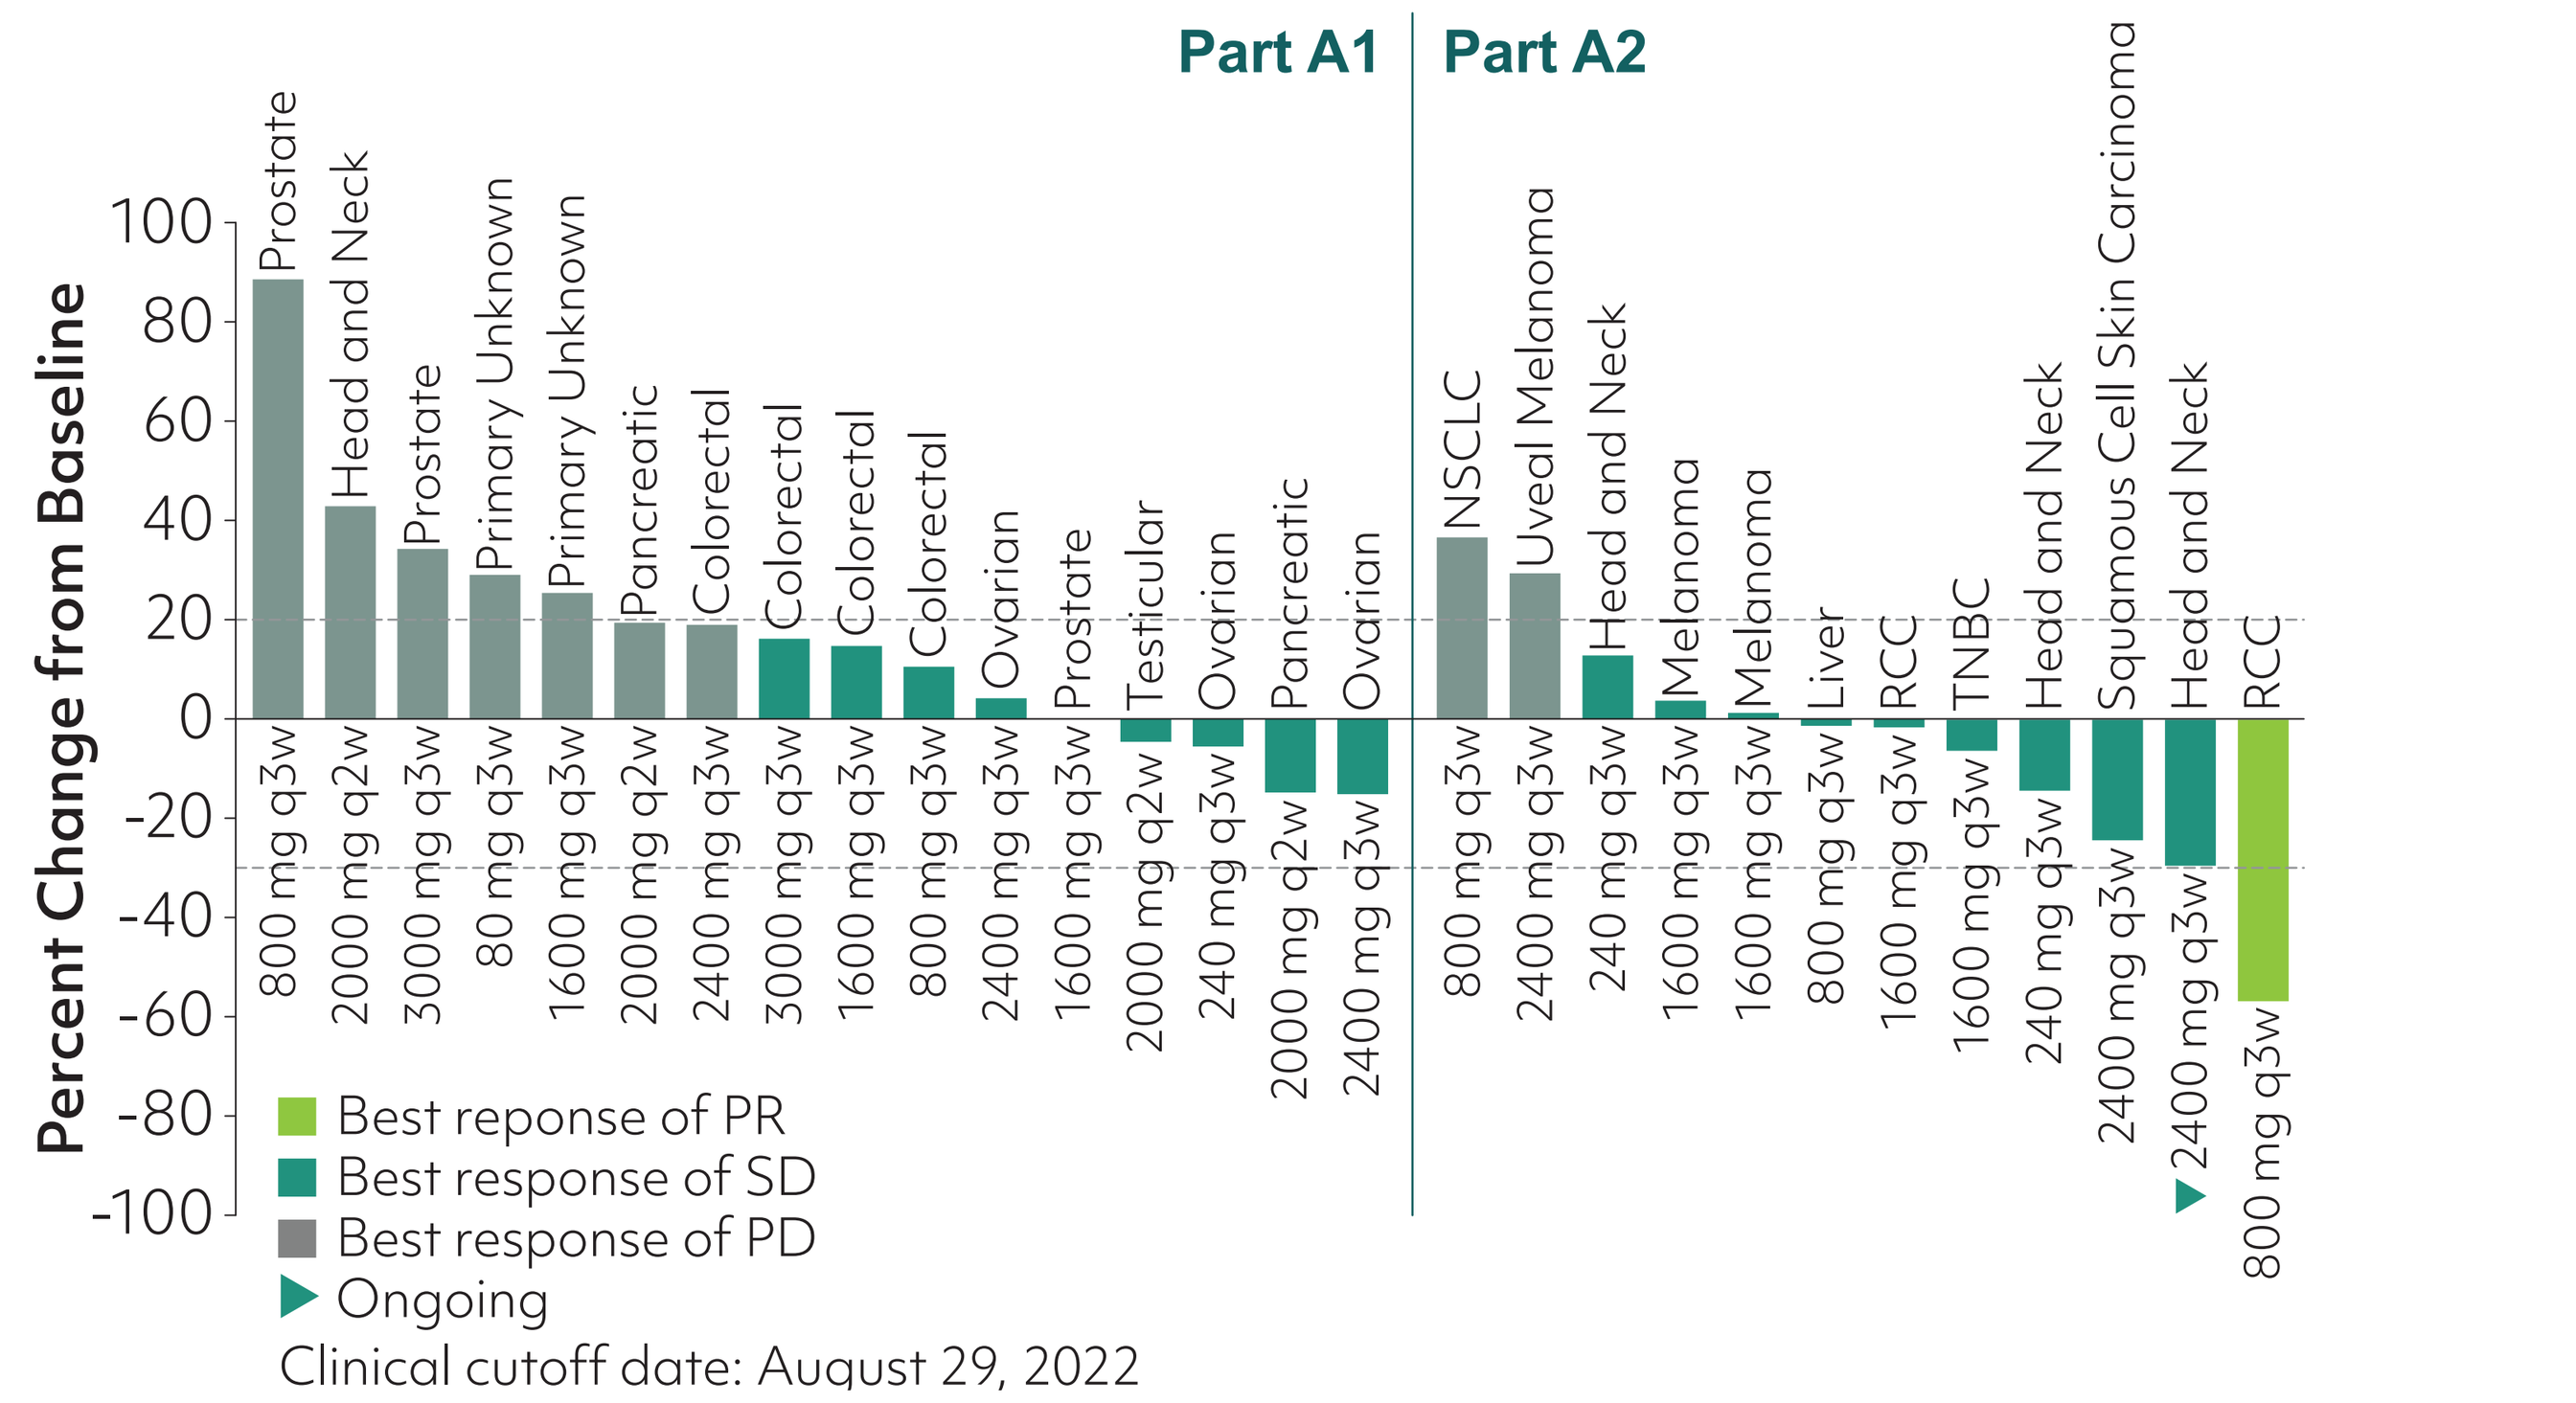
Part A1
Part A2
Percent Change from Baseline
Best reponse of PR
Best response of SD
Best response of PD
Ongoing
Clinical cutoff date: August 29, 2022

Response is assessed using RECIST v1.1 by PI; the scan is performed during screening, 6 weeks after first dose, every 9 weeks for the next 6 months of treatment, and every 12 weeks thereafter.

References: 1) Carretero-González, et al. *Oncotarget*. 2018;9:8706-8715. 2) Mariathasan, et al. *Nature*. 2018;554:544-548. 3) Hugo, et al. *Cell*. 2017;168:542. 4) Martin, et al. *Sci Transl Med*. 2020;12:eaay8456. 5) Brueckner, et al, *Cancer Res*. 2021, 81 (13_Supplement): 1801. 6) Anderton, et al. *Tox Pathol*. 2011;39:916. 7) Stauber, et al. *J Clin Pract*. 2014;4:3. 8) Mitra, et al. *Toxicol Sci*. 2020;175(1):24. 9) Welsh, et al, *Int J Toxicol*. 2021;40(3):226-241. doi:10.1177/1091581821998945. 10) Hoerres, et al. ACCP 2022 Poster 015; doi.org/10.1002/cpdd.835. 11) Yap, et al. *JITC*. 2021;9:doi: 10.1136/jitc-2021-SITC2021.532. 12) Feun, et al. *Cancer*. 2019; 15;125(20):3603-3614.

PK Summary of SRK-181 and Recommended Dose for Part B

- There were no significant differences in the PK profile of SRK-181 as a single agent or in combination with CPI therapy (e.g., pembrolizumab).[10,11]
 - SRK-181 displayed typical monoclonal antibody PK characteristics; preliminary population PK modeling was conducted and population based individual clearance was 11.7 mL/hr and central volume of distribution was 3393 mL.
 - Target-mediated drug disposition was observed at lowest dose level (80 mg) and was overcome beyond the 240 mg dose. Dose proportional increase in exposure was observed beyond the 240 mg dose level.
 - The preliminary population based individual $T_{1/2}$ of SRK-181 was 15 days.
- The recommended Part B dose of 1500 mg q3w and/or 1000mg q2w was chosen based on clinical PK, safety, and ability to maintain the average concentration of SRK-181 measured at a therapeutically relevant dose in mouse tumor models (~80 µg/mL).[10, 11]

Pharmacodynamic Biomarker Results for Part A: Circulatory TGFβ1

- At baseline (Cycle 1, Day 1, pre-infusion), circulatory TGFβ1 was detected in 22 patients with evaluable results (Mean = 2660 pg/mL, Standard Dev. = 782 pg/mL, Min = 1260 pg/mL, Max = 4100 pg/mL).
- Post-treatment with SRK-181, the circulatory TGFβ1 levels increased in all dose groups, indicating an effect related to treatment with SRK-181. However, there was no clear dose dependence apparent in this limited data set.
- The post-treatment increase in circulatory TGFβ1 (fold-change from baseline values) for the combined Q3W dose groups is shown in the figure below.
- Circulatory TGFβ1 levels were similar in Part A1 and A2 patients based on the limited data. Additionally, the combination treatment with pembrolizumab did not appear to impact circulatory TGFβ1 levels.

Median Circulatory TGFβ1 Increased Post-treatment with SRK-181 (Q3W, All Patients)

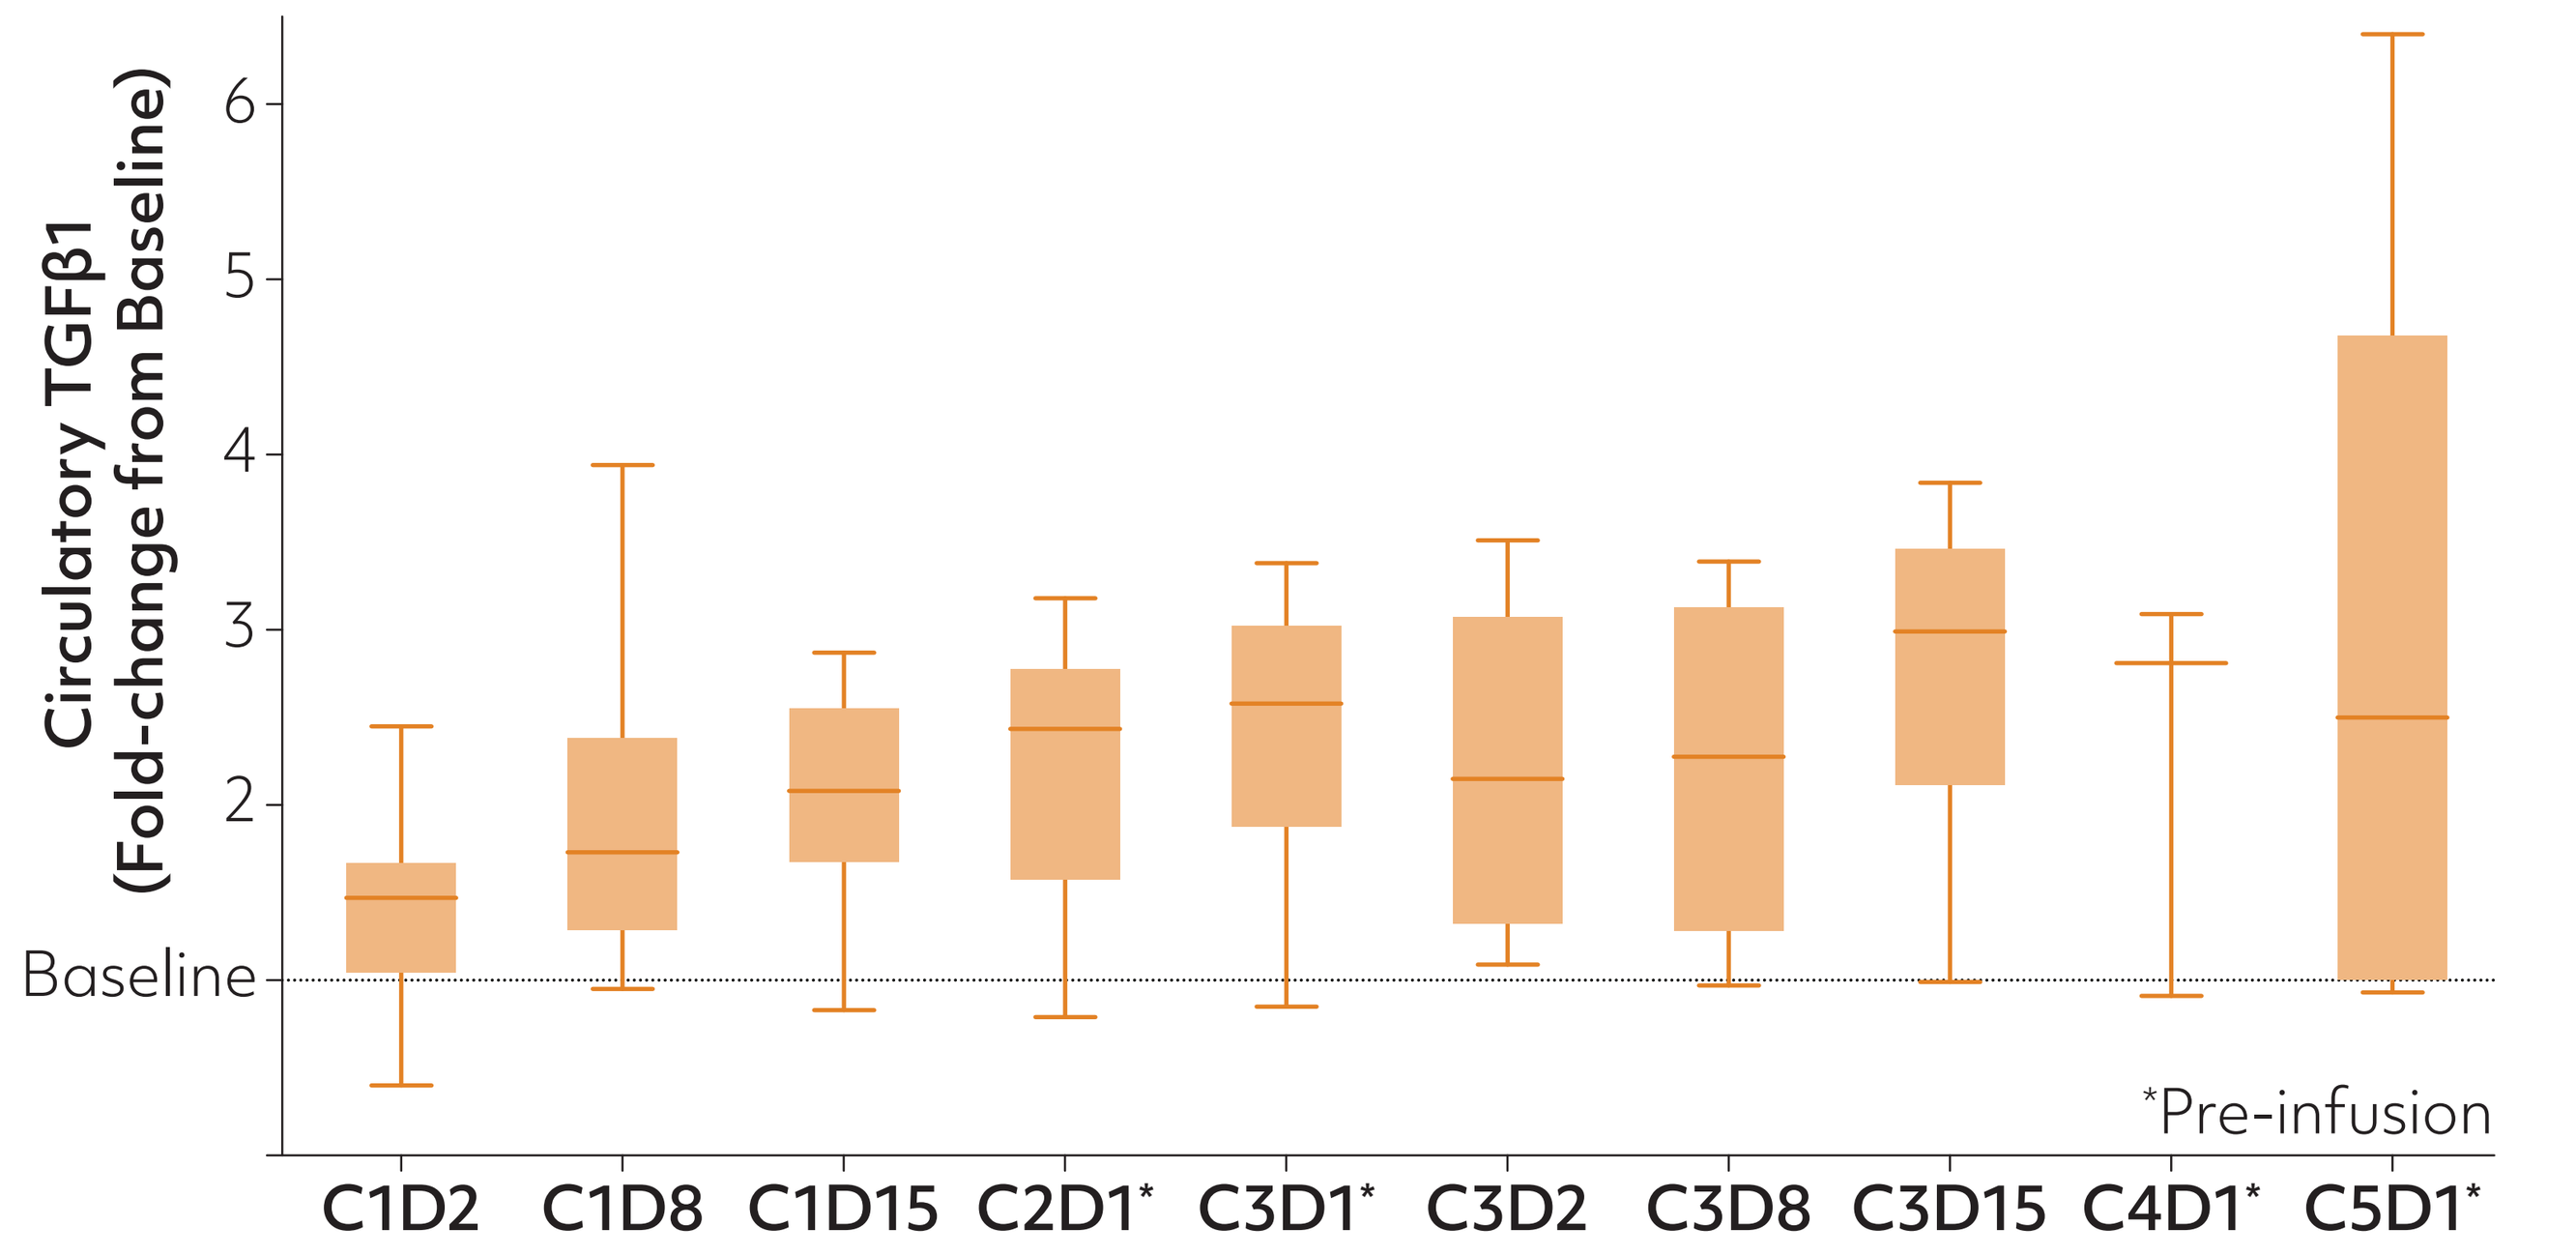
Circulatory TGFβ1 (Fold-change from Baseline)
Baseline
*Pre-infusion
C1D2
C1D8
C1D15
C2D1*
C3D1*
C3D2
C3D8
C3D15
C4D1*
C5D1*

Circulatory TGFβ1 and PF4 levels were quantitated by using validated ELISA kits from R&D System.[12] Because platelet activation during sample processing can lead to elevated TGFβ1 levels, samples with elevated PF4, a platelet activation biomarker, were excluded from the analysis based on a preliminary cutoff value.

Part B Status

- The study enrollment is ongoing.
- As of Aug 29, 2022, 14 patients have been dosed in Part B:
 - The recommended dose of 1500mg q3w or 1000mg q2w SRK-181 in combination with anti-PD-(L)1 was well-tolerated.
 - One confirmed RECIST1.1 PR was observed in a patient with anti-PD-1-resistant ccRCC.

Summary (As of Aug 29, 2022)

- SRK-181 has been well-tolerated both as monotherapy and in combination with anti-PD-(L)1.
- No DLTs were observed up to 3000 mg q3w and 2000 mg q2w in Part A1 and up to 2400 mg q3w in Part A2. The recommended Part B dose is 1500 mg q3w or 1000 mg q2w of SRK-181 + anti-PD-(L)1.
- 2 confirmed PRs were observed in 2 anti-PD-1-resistant ccRCC patients at 800 mg (Part A2) and 1500 mg SRK-181 (Part B) in combination with pembrolizumab. One ongoing patient with head and neck cancer had a tumor reduction of 29.4% at 2400 mg SRK-181 + pembrolizumab (Part A2).

Disclosures: SRK-181 is an investigational drug candidate that is currently being evaluated in a Phase 1 clinical trial. The safety and efficacy of SRK-181 have not been established. SRK-181 has not been approved by the U.S. Food and Drug Administration or any other health authority for any indication.

ScholarRock
medicalinquiry@ScholarRock.com